A4

Negative hyperselection of patients with *RAS* wild-type metastatic colorectal cancer for panitumumab: A biomarker study of the phase III PARADIGM trial

Kohei Shitara,[1] Kei Muro,[2] Jun Watanabe,[3] Kentaro Yamazaki,[4] Hisatsugu Ohori,[5] Manabu Shiozawa,[6] Hirofumi Yasui,[4] Eiji Oki,[7] Takeo Sato,[8] Takeshi Naito,[9] Yoshito Komatsu,[10] Takeshi Kato,[11] Kazunori Yamanaka,[12] Junpei Soeda,[13] Ikuo Mori,[13] Masamitsu Hihara,[13] Kouji Yamamoto,[14] Riu Yamashita,[15] Kiwamu Akagi,[16] Atsushi Ochiai,[17] Hiroyuki Uetake,[18] Katsuya Tsuchihara,[15] Takayuki Yoshino[1]

[1]Department of Gastroenterology and Gastrointestinal Oncology, National Cancer Center Hospital East, Kashiwa, Japan; [2]Department of Clinical Oncology, Aichi Cancer Center Hospital, Nagoya, Japan; [3]Gastroenterological Center, Yokohama City University Medical Center, Yokohama, Japan; [4]Division of Gastrointestinal Oncology, Shizuoka Cancer Center, Shizuoka, Japan; [5]Division of Medical Oncology, Japanese Red Cross Ishinomaki Hospital, Miyagi, Japan; [6]Division of Gastrointestinal Surgery, Kanagawa Cancer Center, Kanagawa, Japan; [7]Department of Surgery and Science, Graduate School of Medical Sciences, Kyushu University, Fukuoka, Japan; [8]Research and Development Center for Medical Education, Department of Clinical Skills Education, Kitasato University School of Medicine, Sagamihara, Japan; [9]Department of Lower Gastrointestinal Surgery, Kitasato University School of Medicine, Sagamihara, Japan; [10]Division of Cancer Chemotherapy, Hokkaido University Hospital Cancer Center, Sapporo, Japan; [11]Department of Surgery, National Hospital Organization Osaka National Hospital, Osaka, Japan; [12]Pharmaceutical Research Division, Takeda Pharmaceutical Company Ltd, Tokyo, Japan; [13]Japan Medical Affairs, Japan Oncology Business Unit, Takeda Pharmaceutical Company Ltd, Tokyo, Japan; [14]Department of Biostatistics, Yokohama City University School of Medicine, Yokohama, Japan; [15]Division of Translational Informatics, Exploratory Oncology Research & Clinical Trial Center, National Cancer Center, Chiba, Japan; [16]Division of Molecular Diagnosis and Cancer Prevention, Saitama Cancer Center, Saitama, Japan; [17]Research Institute for Biomedical Sciences, Tokyo University of Science, Tokyo, Japan; [18]National Hospital Organization, Disaster Medical Center, Tokyo, Japan

For an e-Print, scan this QR code.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the authors of this poster.

Introduction

- The phase 3 PARADIGM trial (NCT02394795) met the primary endpoint, demonstrating the superiority of first-line panitumumab in combination with modified FOLFOX6 (mFOLFOX6) vs bevacizumab in combination with mFOLFOX6 in the left-sided and overall *RAS* wild-type (WT) metastatic colorectal cancer (mCRC) populations[1]
 - Left-sided population: median overall survival (mOS), 37.9 vs 34.3 months; hazard ratio (HR), 0.82 (95.798% CI: 0.68–0.99); *P*=0.031
 - Overall population: mOS, 36.2 vs 31.3 months; HR, 0.84 (95% CI: 0.72–0.98); *P*=0.030
 - Right-sided population (exploratory endpoint): mOS, 20.2 vs 23.2 months; HR, 1.09 (95% CI: 0.79–1.51)
- The PARADIGM biomarker study (NCT02394834) was designed to investigate potential biomarkers related to primary and secondary resistance of each therapy by using tumor tissue and circulating tumor DNA (ctDNA)
- In this analysis, we evaluated the usefulness of negative hyperselection by gene alterations in ctDNA related to primary resistance to anti-epidermal growth factor receptor (EGFR) therapy in patients enrolled in the PARADIGM biomarker study

Methods

Figure 1: Study design

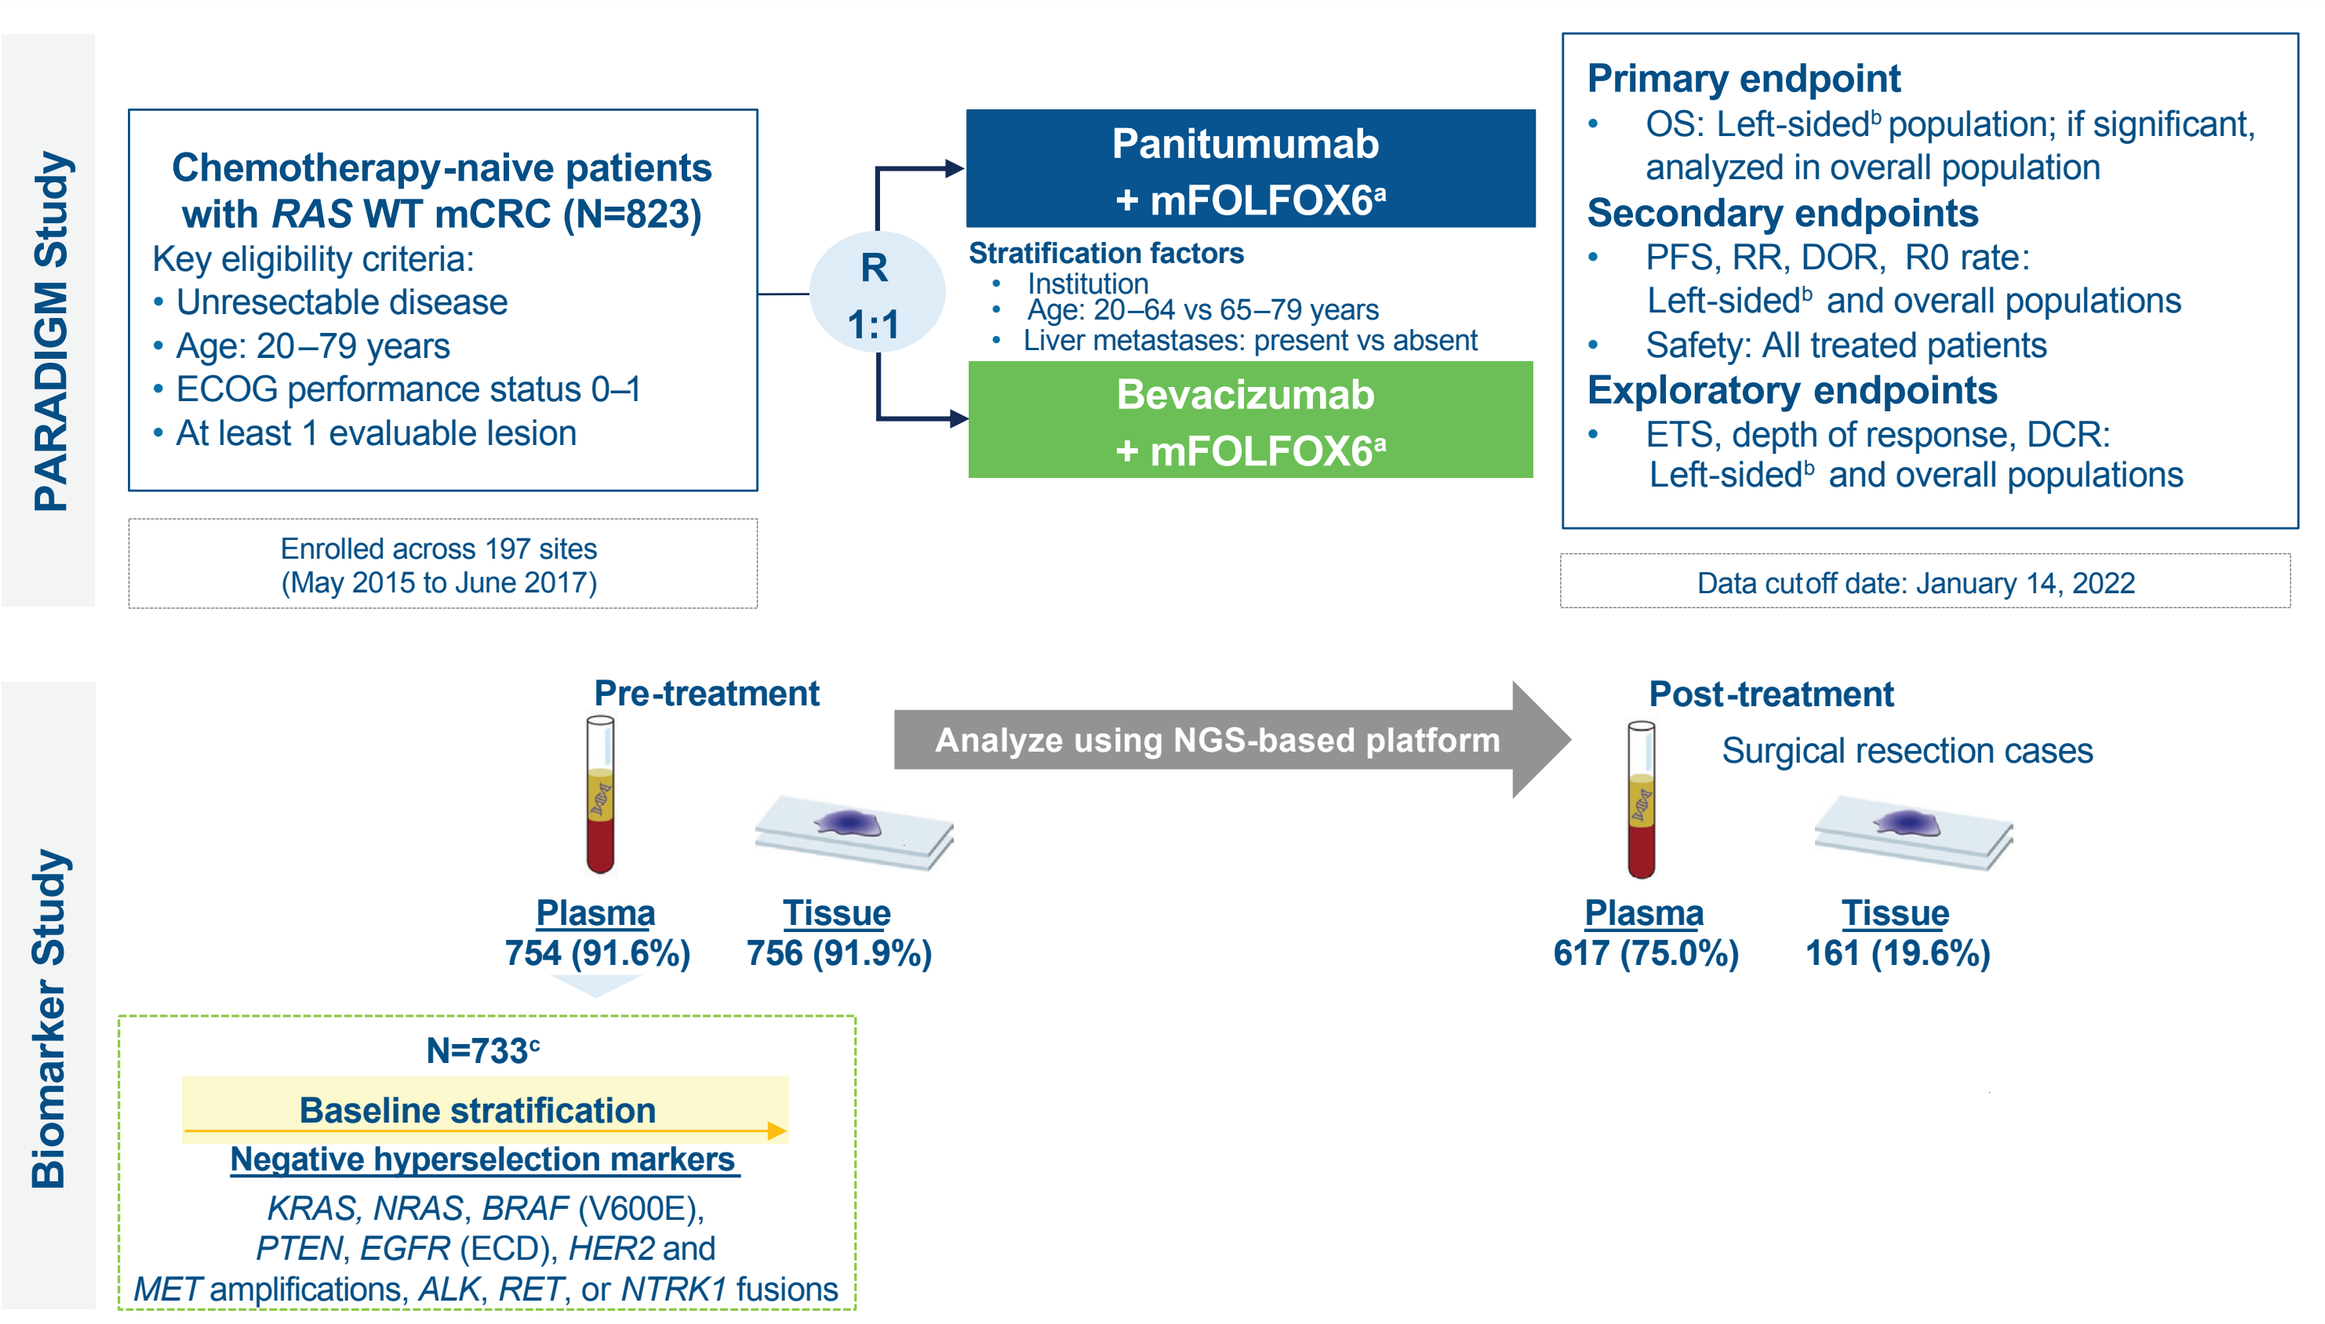
PARADIGM Study
Chemotherapy-naive patients with RAS WT mCRC (N=823)
Key eligibility criteria:
• Unresectable disease
• Age: 20–79 years
• ECOG performance status 0–1
• At least 1 evaluable lesion
Enrolled across 197 sites (May 2015 to June 2017)
R 1:1
Panitumumab + mFOLFOX6a
Stratification factors
Institution
Age: 20–64 vs 65–79 years
Liver metastases: present vs absent
Bevacizumab + mFOLFOX6a
Primary endpoint
OS: Left-sidedb population; if significant, analyzed in overall population
Secondary endpoints
PFS, RR, DOR, R0 rate: Left-sidedb and overall populations
Safety: All treated patients
Exploratory endpoints
ETS, depth of response, DCR: Left-sidedb and overall populations
Data cutoff date: January 14, 2022
Biomarker Study
Pre-treatment
Analyze using NGS-based platform
Post-treatment
Surgical resection cases
Plasma 754 (91.6%)
Tissue 756 (91.9%)
Plasma 617 (75.0%)
Tissue 161 (19.6%)
N=733c
Baseline stratification
Negative hyperselection markers
KRAS, NRAS, BRAF (V600E), PTEN, EGFR (ECD), HER2 and MET amplifications, ALK, RET, or NTRK1 fusions

[a]Until disease progression, unacceptable toxicity, withdrawal of consent, investigator's judgement, or curative intent resection; [b]Primary tumor in descending colon, sigmoid colon, rectosigmoid, and rectum; [c]Patients with available ctDNA among those included in efficacy analysis set in the PARADIGM study
ALK, anaplastic lymphoma kinase; Amp, amplification; *BRAF*, v-raf murine sarcoma viral oncogene homolog B1; DCR, disease control rate; DOR, duration of response; ECD, extracellular domain; ECOG, Eastern Cooperative Oncology Group; ETS, early tumor shrinkage; *HER2*, human epidermal growth factor receptor 2; *MET*, mesenchymal epithelial transition factor receptor; NGS, next-generation sequencing; *NTRK1*, neurotrophic tyrosine receptor kinase 1; PFS, progression-free survival; *PTEN*, phosphatase and tensin homolog; R, randomization; R0, margin-negative resection; *RET*, rearranged during transfection; RR, response rate; WT, wild-type

- Baseline plasma ctDNA (>10 ng/mL and >10 nM DNA) from patients enrolled in the biomarker study was assessed using a custom panel (PlasmaSELECT-R 91, PGDx)
- The panel was designed to detect mutations, amplifications, and rearrangements in 90, 26, and 3 mCRC-related genes, respectively, as well as microsatellite instability
 - Targeted genomic regions spanned 250 kb
- Prespecified gene alterations for hyperselection that have been reported to confer resistance to anti-EGFR antibody therapy included *KRAS*, *NRAS*, *BRAF* (V600E), *PTEN*, and extracellular domain *EGFR* mutations, *HER2* and *MET* amplifications, and *ALK*, *RET*, or *NTRK1* fusions[2-5]
- Hyperselection status (all negative vs gene altered [any positive biomarker]) was correlated with OS, PFS, and RR in the PARADIGM study population

Results

- ctDNA status was evaluable in 91% (733/802) of patients (**Figure 2**; **Figure 3**)
 - 28% of patients had at least 1 gene alteration
 - 72% of patients had no gene alterations and were classified as hyperselected patients
- When stratified by primary tumor sidedness, any gene alteration was detected in 21% of patients with left-sided mCRC and 50% of patients with right-sided mCRC

Figure 2: Disposition

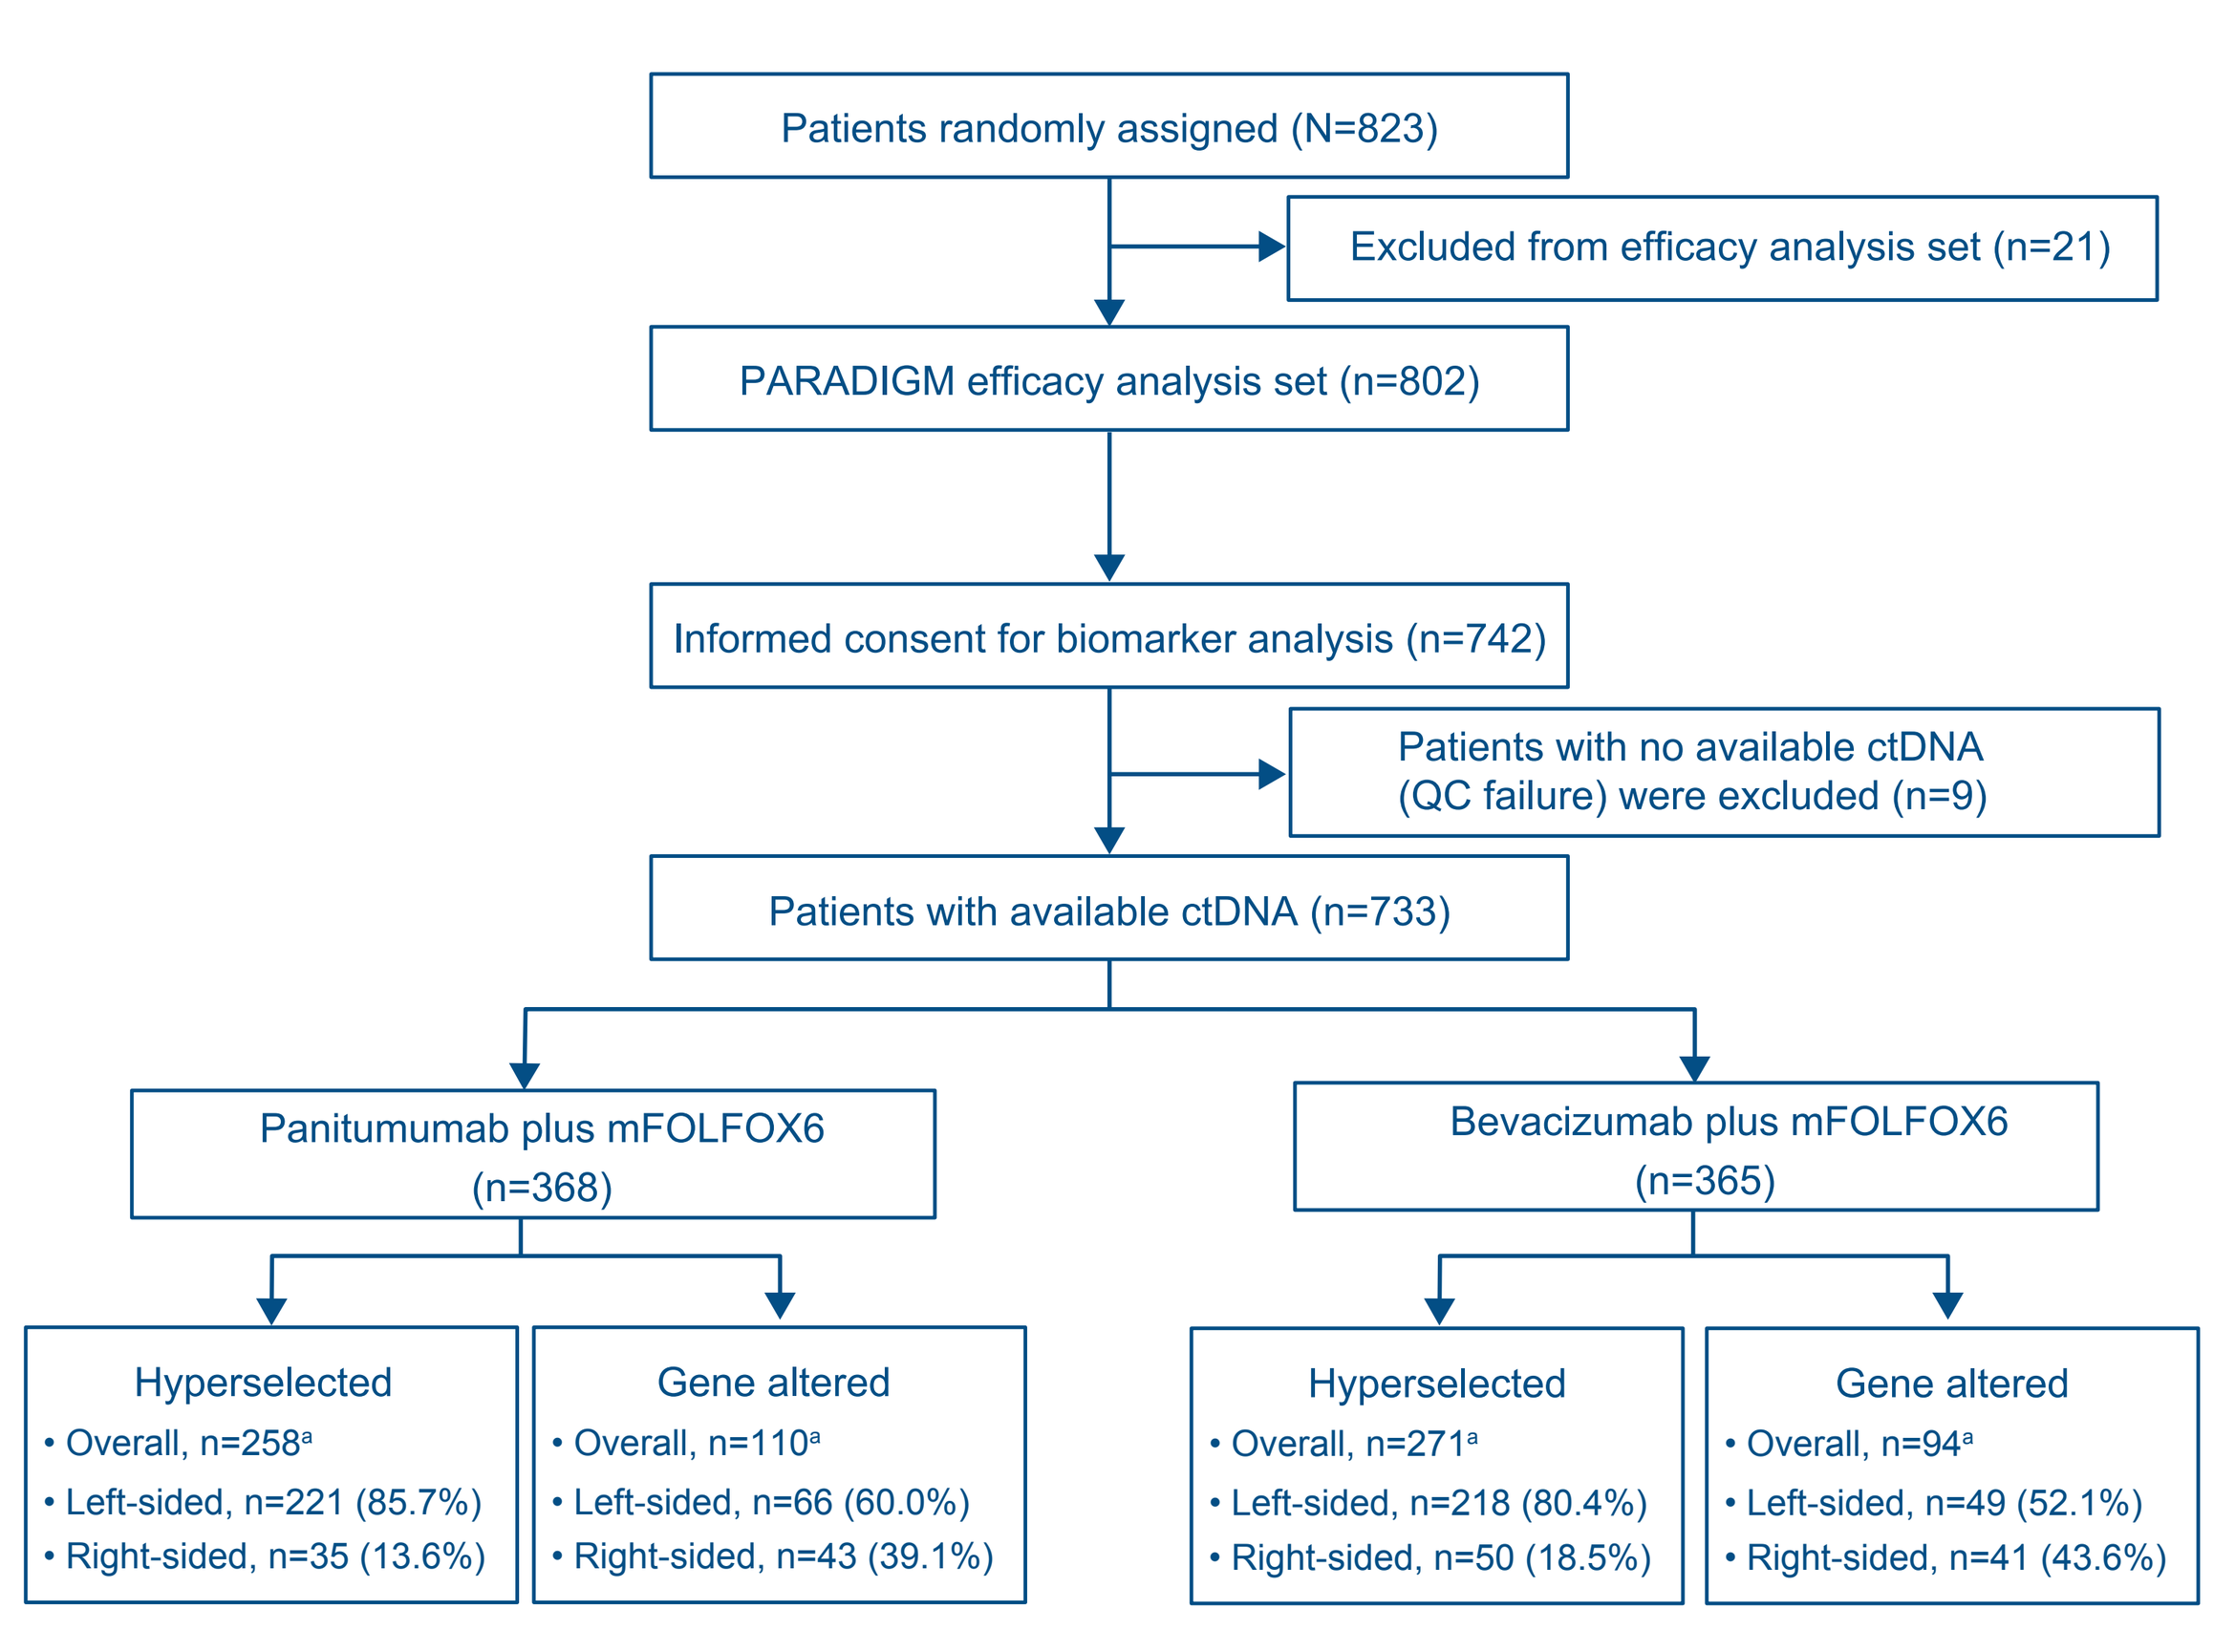
Patients randomly assigned (N=823)
Excluded from efficacy analysis set (n=21)
PARADIGM efficacy analysis set (n=802)
Informed consent for biomarker analysis (n=742)
Patients with no available ctDNA (QC failure) were excluded (n=9)
Patients with available ctDNA (n=733)
Panitumumab plus mFOLFOX6 (n=368)
Bevacizumab plus mFOLFOX6 (n=365)
Hyperselected
• Overall, n=258a
• Left-sided, n=221 (85.7%)
• Right-sided, n=35 (13.6%)
Gene altered
• Overall, n=110a
• Left-sided, n=66 (60.0%)
• Right-sided, n=43 (39.1%)
Hyperselected
• Overall, n=271a
• Left-sided, n=218 (80.4%)
• Right-sided, n=50 (18.5%)
Gene altered
• Overall, n=94a
• Left-sided, n=49 (52.1%)
• Right-sided, n=41 (43.6%)

[a]Some patients had multiple primary lesions in both the left and right sides
QC, quality control

- Baseline characteristics of patients who were hyperselected or had gene alteration within each treatment arm and across left- and right-sided mCRC populations are shown in **Table 1**

Table 1: Baseline patient characteristics

Characteristic	Overall population (N=733)				Left-sided population (N=554)				Right-sided population (N=169)			
	Hyperselected		Gene Altered		Hyperselected		Gene Altered		Hyperselected		Gene Altered	
	Panitumumab +mFOLFOX6 n=258	Bevacizumab +mFOLFOX6 n=271	Panitumumab +mFOLFOX6 n=110	Bevacizumab +mFOLFOX6 n=94	Panitumumab +mFOLFOX6 n=221	Bevacizumab +mFOLFOX6 n=218	Panitumumab +mFOLFOX6 n=66	Bevacizumab +mFOLFOX6 n=49	Panitumumab +mFOLFOX6 n=35	Bevacizumab +mFOLFOX6 n=50	Panitumumab +mFOLFOX6 n=43	Bevacizumab +mFOLFOX6 n=41
Age category, n (%)												
20–64 years	104 (40.3)	116 (42.8)	45 (40.9)	36 (38.3)	94 (42.5)	91 (41.7)	30 (45.5)	23 (46.9)	10 (28.6)	24 (48.0)	15 (34.9)	12 (29.3)
65–79 years	154 (59.7)	155 (57.2)	65 (59.1)	58 (61.7)	127 (57.5)	127 (58.3)	36 (54.5)	26 (53.1)	25 (71.4)	26 (52.0)	28 (65.1)	29 (70.7)
Sex, female, n (%)	87 (33.7)	83 (30.6)	47 (42.7)	37 (39.4)	71 (32.1)	66 (30.3)	23 (34.8)	15 (30.6)	16 (45.7)	17 (34.0)	23 (53.5)	21 (51.2)
ECOG performance status, n (%)												
0	219 (84.9)	213 (78.6)	85 (77.3)	75 (80.0)	187 (84.6)	171 (78.4)	54 (81.8)	39 (79.6)	31 (88.6)	39 (78.0)	31 (72.1)	33 (80.5)
1	39 (15.1)	58 (21.4)	24 (21.8)	19 (20.2)	34 (15.4)	47 (21.6)	12 (18.2)	10 (20.4)	4 (11.4)	11 (22.0)	11 (25.6)	8 (19.5)
Primary tumor location, n (%)												
Left-sided	221 (85.7)	218 (80.4)	66 (60.0)	49 (52.1)	221 (100)	218 (100)	66 (100)	49 (100)	0	0	0	0
Right-sided	35 (13.6)	50 (18.5)	43 (39.1)	41 (43.6)	0	0	0	0	35 (100)	50 (100)	43 (100)	41 (100)
Number of metastatic organs, n (%)												
1	141 (54.7)	139 (51.3)	40 (36.4)	39 (41.5)	120 (54.3)	115 (52.8)	21 (31.8)	20 (40.8)	20 (57.1)	22 (44.0)	19 (44.2)	18 (43.9)
≥2	117 (45.3)	132 (48.7)	70 (63.6)	55 (58.5)	101 (45.7)	103 (47.2)	45 (68.2)	29 (59.2)	15 (42.9)	28 (56.0)	24 (55.8)	23 (56.1)
Metastatic site, n (%)												
Liver	172 (66.7)	182 (67.2)	82 (74.5)	66 (70.2)	153 (69.2)	147 (67.4)	55 (83.3)	58 (77.6)	18 (51.4)	32 (64.0)	27 (62.8)	25 (61.0)
Liver as only site of metastasis	73 (28.3)	78 (28.8)	23 (20.9)	24 (25.5)	66 (29.9)	64 (29.4)	16 (24.2)	16 (32.7)	6 (17.1)	12 (24.0)	7 (16.3)	7 (17.1)
Prior treatment, n (%)												
Primary tumor resection	165 (64.0)	184 (67.9)	57 (51.8)	60 (63.8)	136 (61.5)	145 (66.5)	35 (53.0)	30 (61.2)	27 (77.1)	36 (72.0)	22 (51.2)	27 (65.9)

- The OncoPrint profile of mutational frequencies and types of alterations of the 10 genes sequenced are shown in **Figure 3**

Figure 3: Co-occurring gene alterations in left- and right-sided tumors

	Left-sided (n=554)	Right-sided (n=169)	Other (n=10)	Overall (N=733)
Gene altered n (%)	115 (20.8)	84 (49.7)	5 (50)	204 (27.8)
Hyperselected n (%)	439 (79.2)	85 (50.3)	5 (50)	529 (72.2)

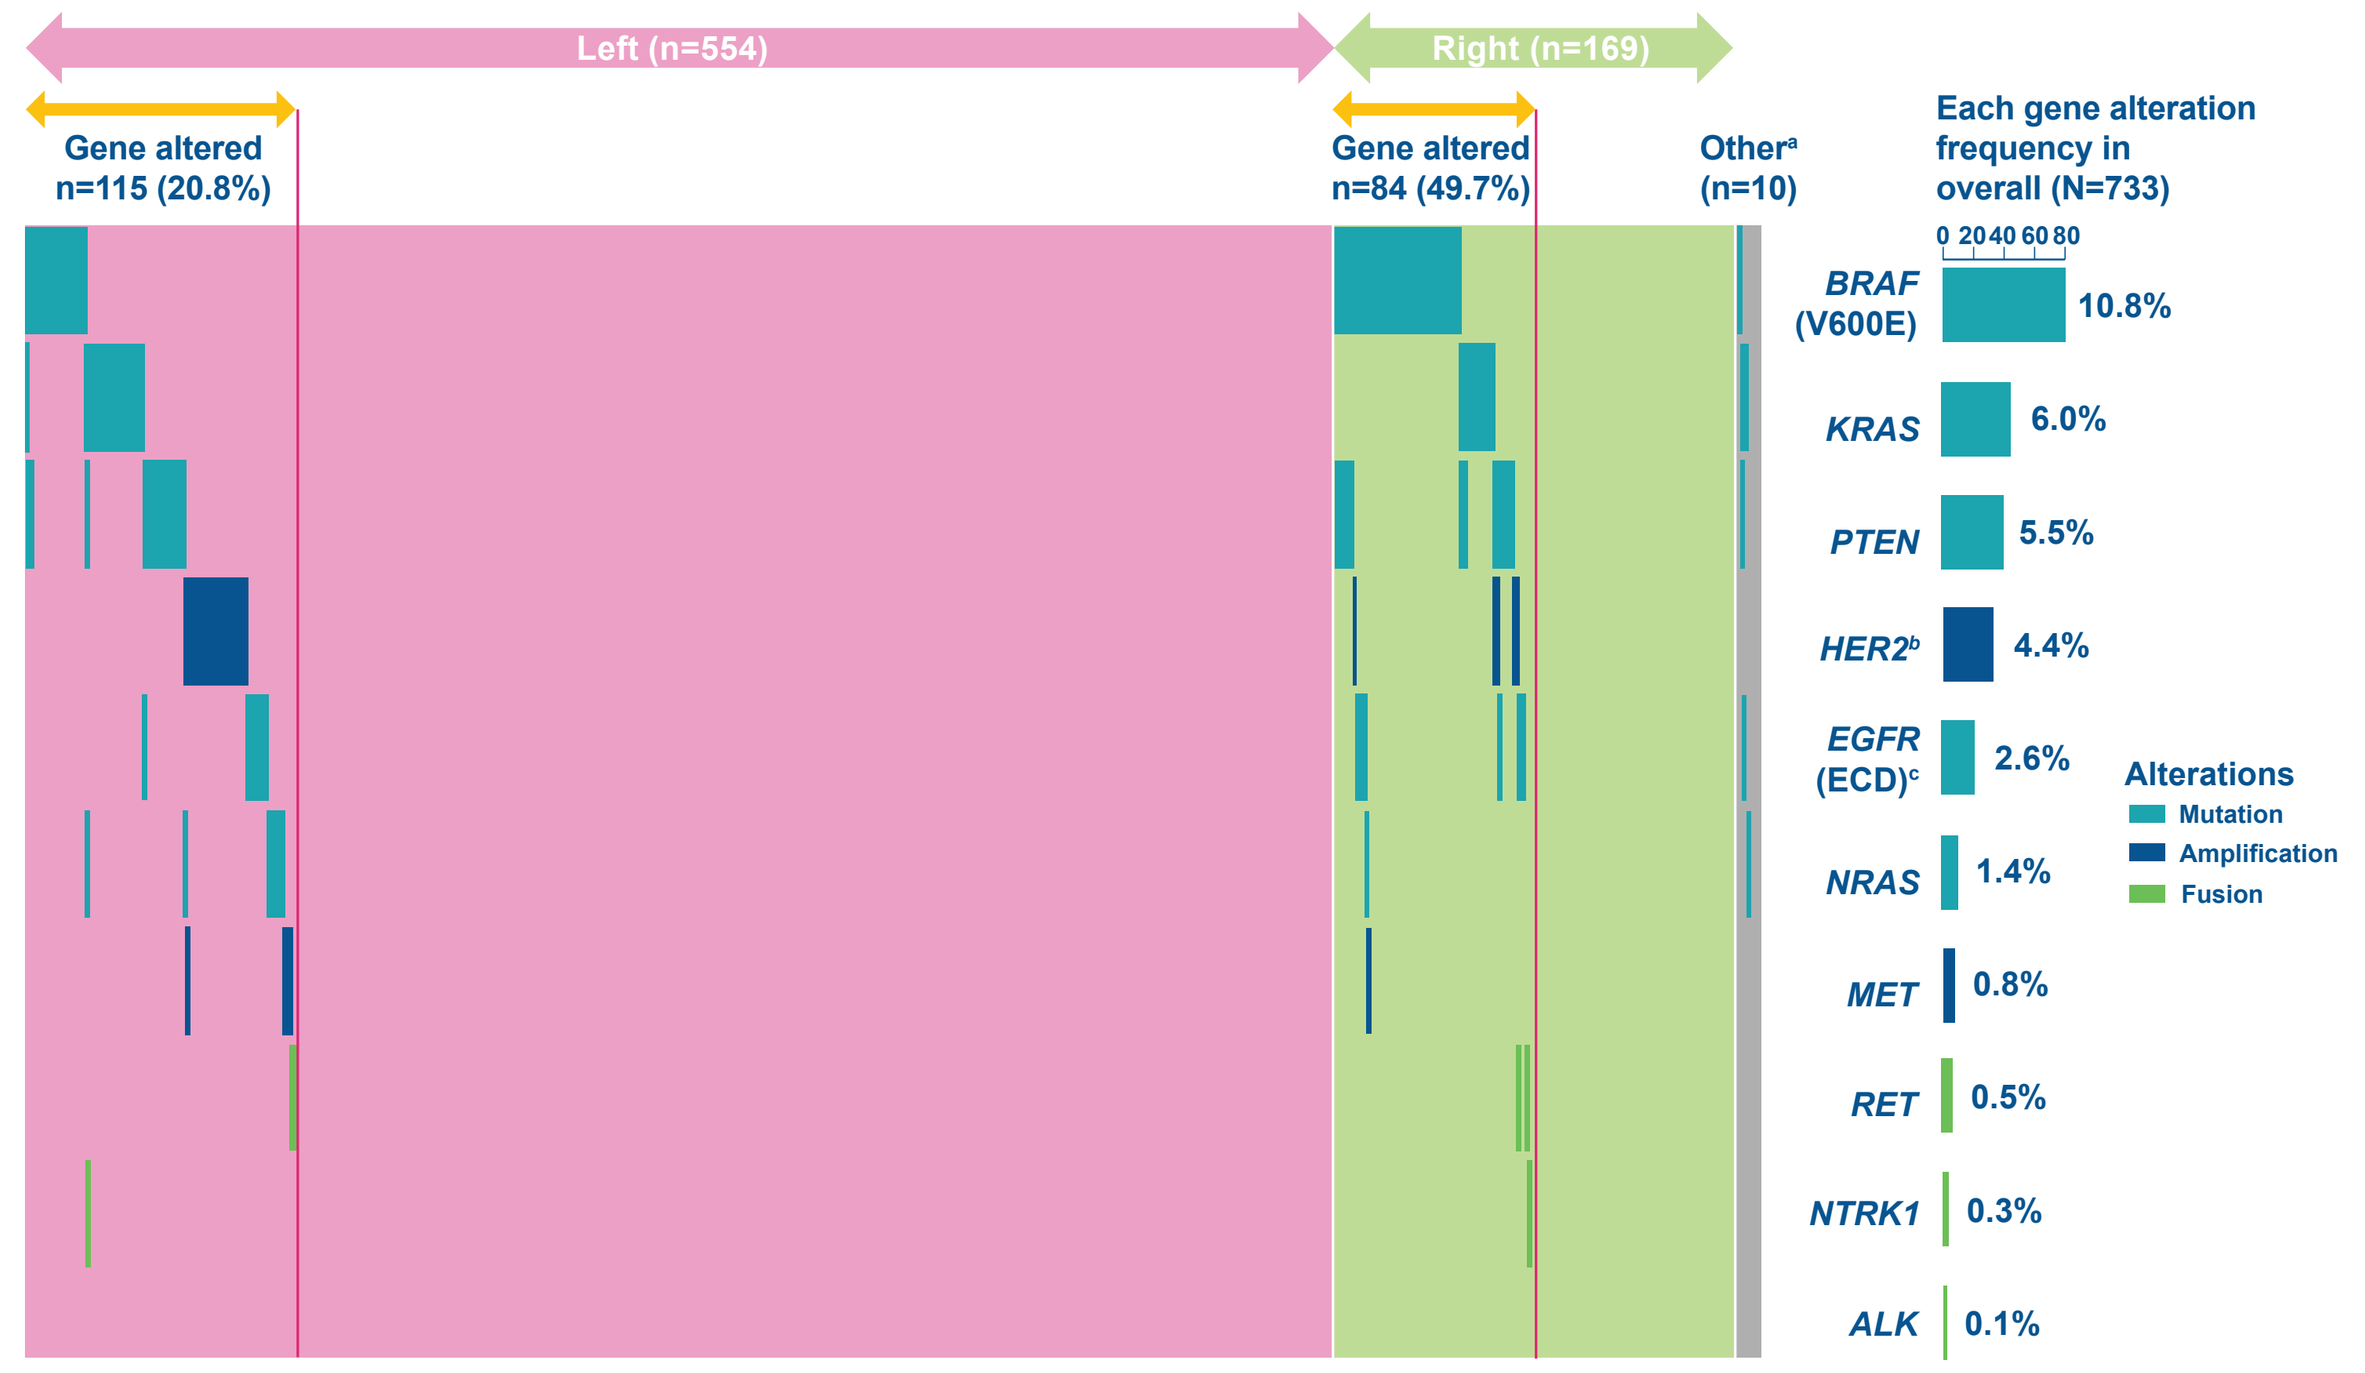
Left (n=554)
Right (n=169)
Gene altered n=115 (20.8%)
Gene altered n=84 (49.7%)
Othera (n=10)
Each gene alteration frequency in overall (N=733)
0 20 40 60 80
BRAF (V600E) 10.8%
KRAS 6.0%
PTEN 5.5%
HER2b 4.4%
EGFR (ECD)c 2.6%
NRAS 1.4%
MET 0.8%
RET 0.5%
NTRK1 0.3%
ALK 0.1%
Alterations
Mutation
Amplification
Fusion

[a]Patients who had multiple primary lesions in both the left and right sides; [b]The custom panel (Tak_Seq3) has a 1.25 threshold for *HER2* (thresholds were set based on noise in normal samples); [c]*EGFR* (ECD): Exon 1–16 (1–620)

Table 2: Number of genetic alterations in ctDNA

Gene alteration, n (%)	Overall population (N=733)		Left-sided population (n=554)		Right-sided population (n=169)	
	Panitumumab (n=368)	Bevacizumab (n=365)	Panitumumab (n=287)	Bevacizumab (n=267)	Panitumumab (n=78)	Bevacizumab (n=91)
BRAF (V600E)	43 (11.7)	36 (9.9)	17 (5.9)	8 (3.0)	26 (33.3)	27 (29.7)
KRAS	22 (6.0)	23 (6.3)	11 (3.8)	15 (5.6)	9 (11.5)	6 (6.6)
PTEN	23 (6.3)	17 (4.7)	12 (4.2)	8 (3.0)	10 (12.8)	9 (9.9)
HER2 amplification	19 (5.2)	14 (3.8)	16 (5.6)	11 (4.1)	3 (3.8)	2 (2.2)
EGFR (ECD)	12 (3.3)	7 (1.9)	7 (2.4)	3 (1.1)	5 (6.4)	3 (3.3)
NRAS	10 (2.7)	3 (0.8)	6 (2.1)	2 (0.7)	1 (1.3)	0
MET amplification	3 (0.8)	2 (0.5)	3 (1.0)	2 (0.7)	0	0
RET fusion	2 (0.5)	2 (0.5)	0	2 (0.7)	2 (2.6)	0
NTRK1 fusion	1 (0.3)	1 (0.3)	0	1 (0.4)	1 (1.3)	0
ALK fusion	0	1 (0.3)	0	0	0	1 (1.1)

Figure 4: Survival outcomes in the overall population analyzed for ctDNA

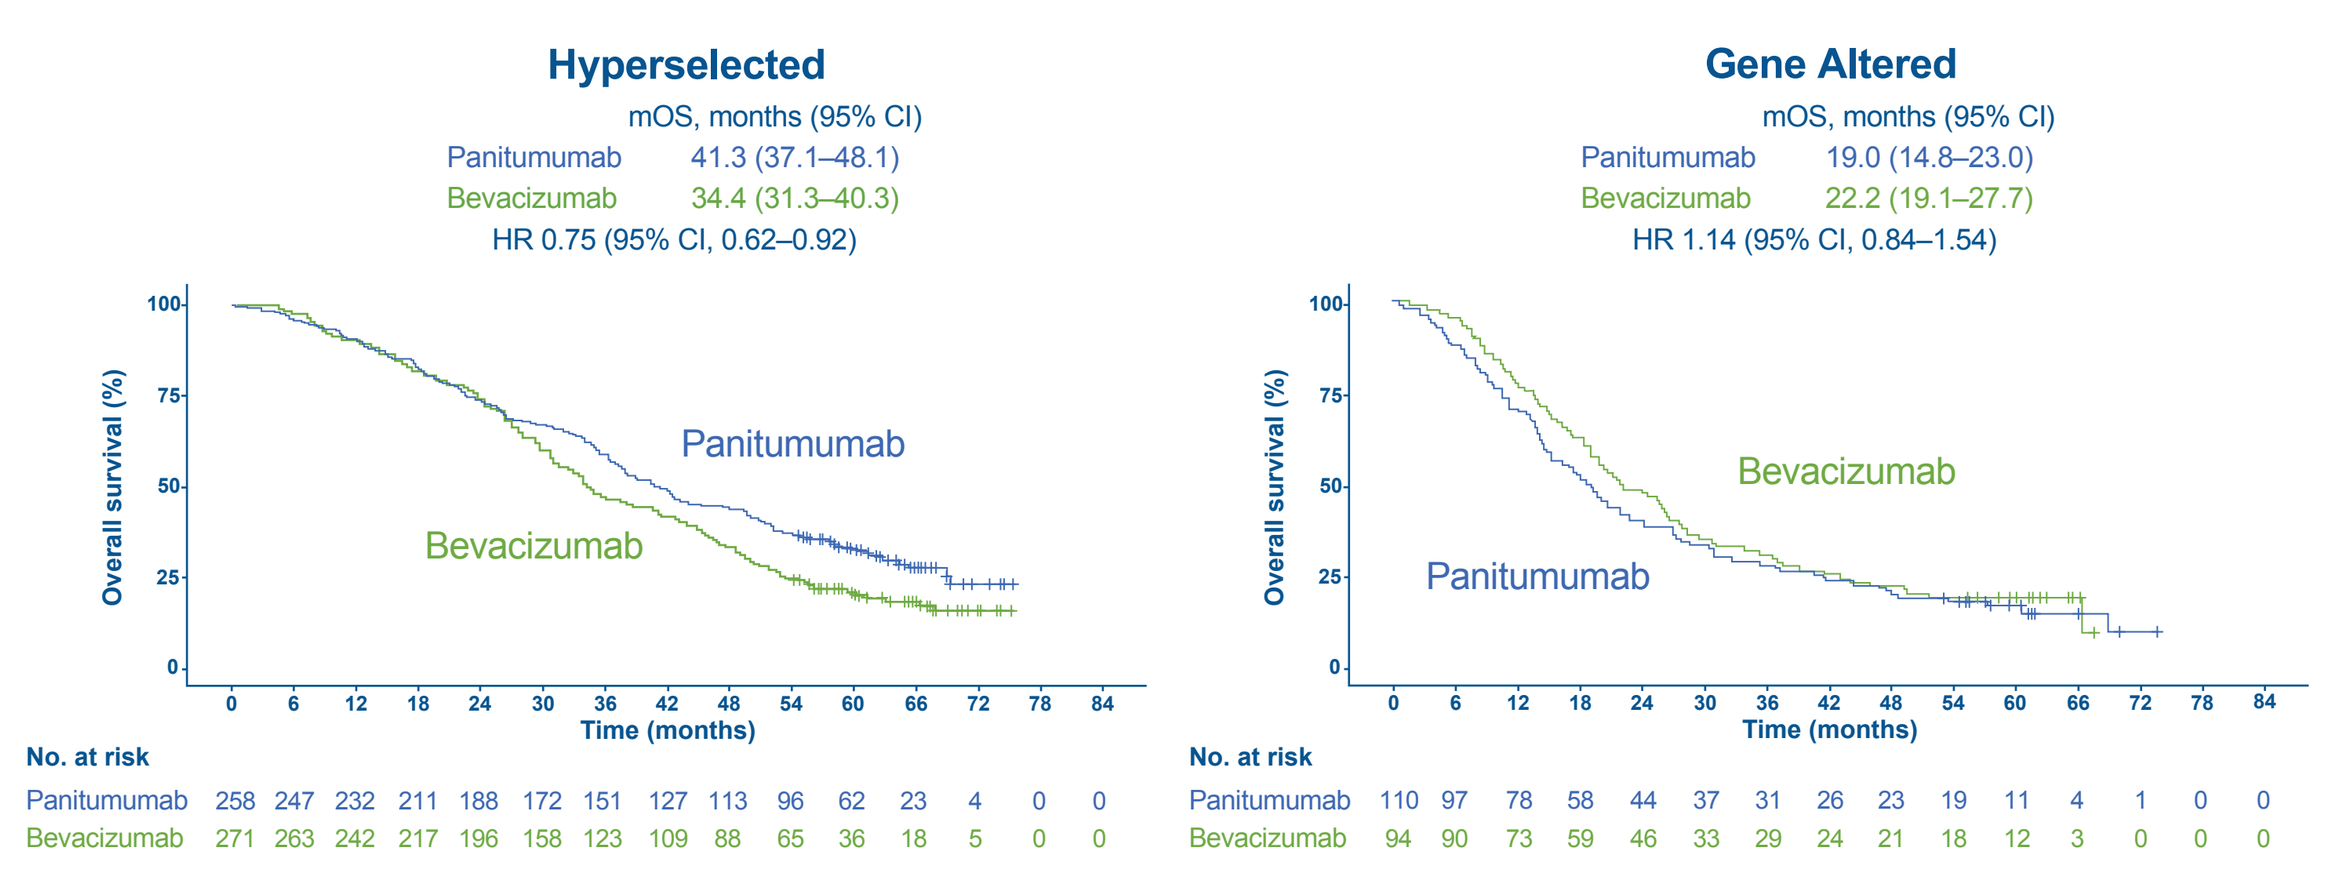
Hyperselected
mOS, months (95% CI)
Panitumumab 41.3 (37.1–48.1)
Bevacizumab 34.4 (31.3–40.3)
HR 0.75 (95% CI, 0.62–0.92)
Gene Altered
mOS, months (95% CI)
Panitumumab 19.0 (14.8–23.0)
Bevacizumab 22.2 (19.1–27.7)
HR 1.14 (95% CI, 0.84–1.54)
Overall survival (%)
Time (months)
Panitumumab
Bevacizumab
No. at risk
Panitumumab 258 247 232 211 188 172 151 127 113 96 62 23 4 0 0
Bevacizumab 271 263 242 217 196 158 123 109 88 65 36 18 5 0 0
No. at risk
Panitumumab 110 97 78 58 44 37 31 26 23 19 11 4 1 0 0
Bevacizumab 94 90 73 59 46 33 29 24 21 18 12 3 0 0 0

Figure 5: Survival outcomes in the left-sided mCRC population analyzed for ctDNA

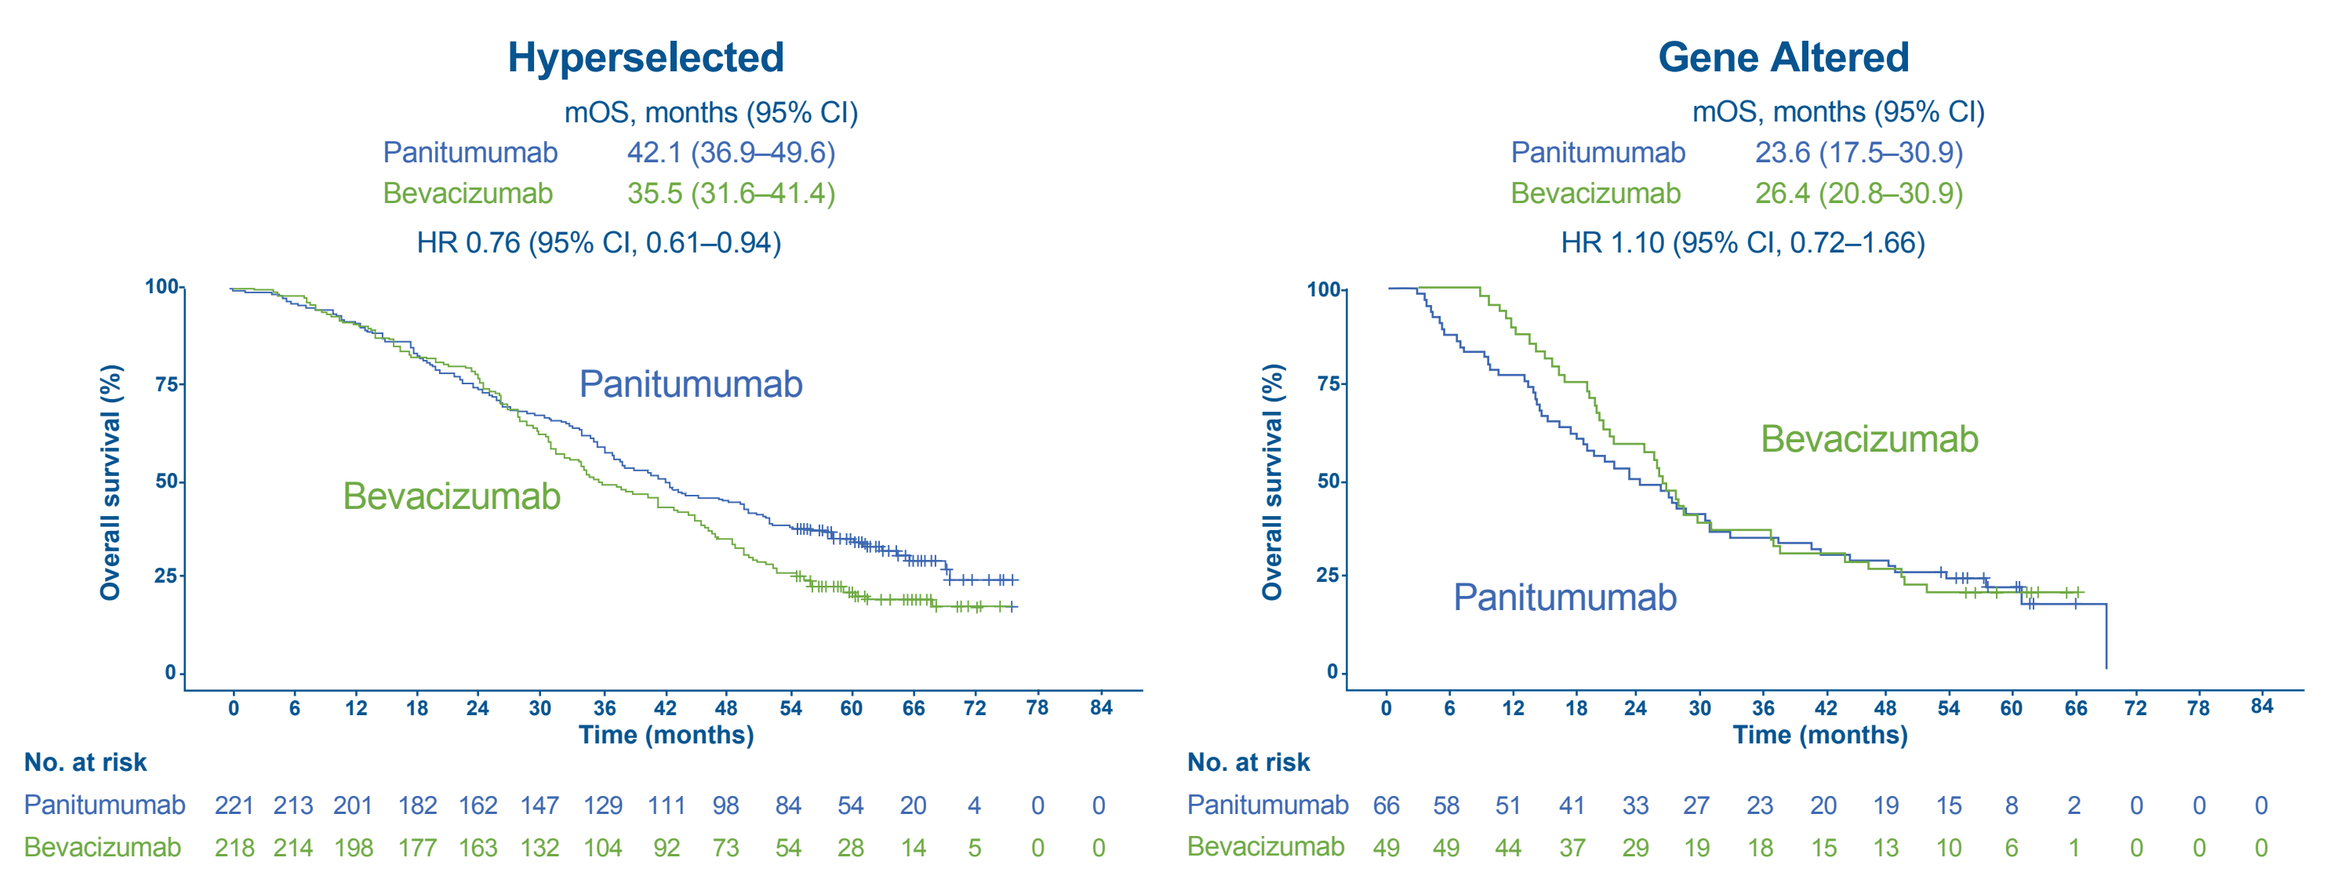
Hyperselected
mOS, months (95% CI)
Panitumumab 42.1 (36.9–49.6)
Bevacizumab 35.5 (31.6–41.4)
HR 0.76 (95% CI, 0.61–0.94)
Gene Altered
mOS, months (95% CI)
Panitumumab 23.6 (17.5–30.9)
Bevacizumab 26.4 (20.8–30.9)
HR 1.10 (95% CI, 0.72–1.66)
Overall survival (%)
Time (months)
Panitumumab
Bevacizumab
No. at risk
Panitumumab 221 213 201 182 162 147 129 111 98 84 54 20 4 0 0
Bevacizumab 218 214 198 177 163 132 104 92 73 54 28 14 5 0 0
No. at risk
Panitumumab 66 58 51 41 33 27 23 20 19 15 8 2 0 0 0
Bevacizumab 49 49 44 37 29 19 18 15 13 10 6 1 0 0 0

Figure 6: Survival outcomes in the right-sided mCRC population analyzed for ctDNA

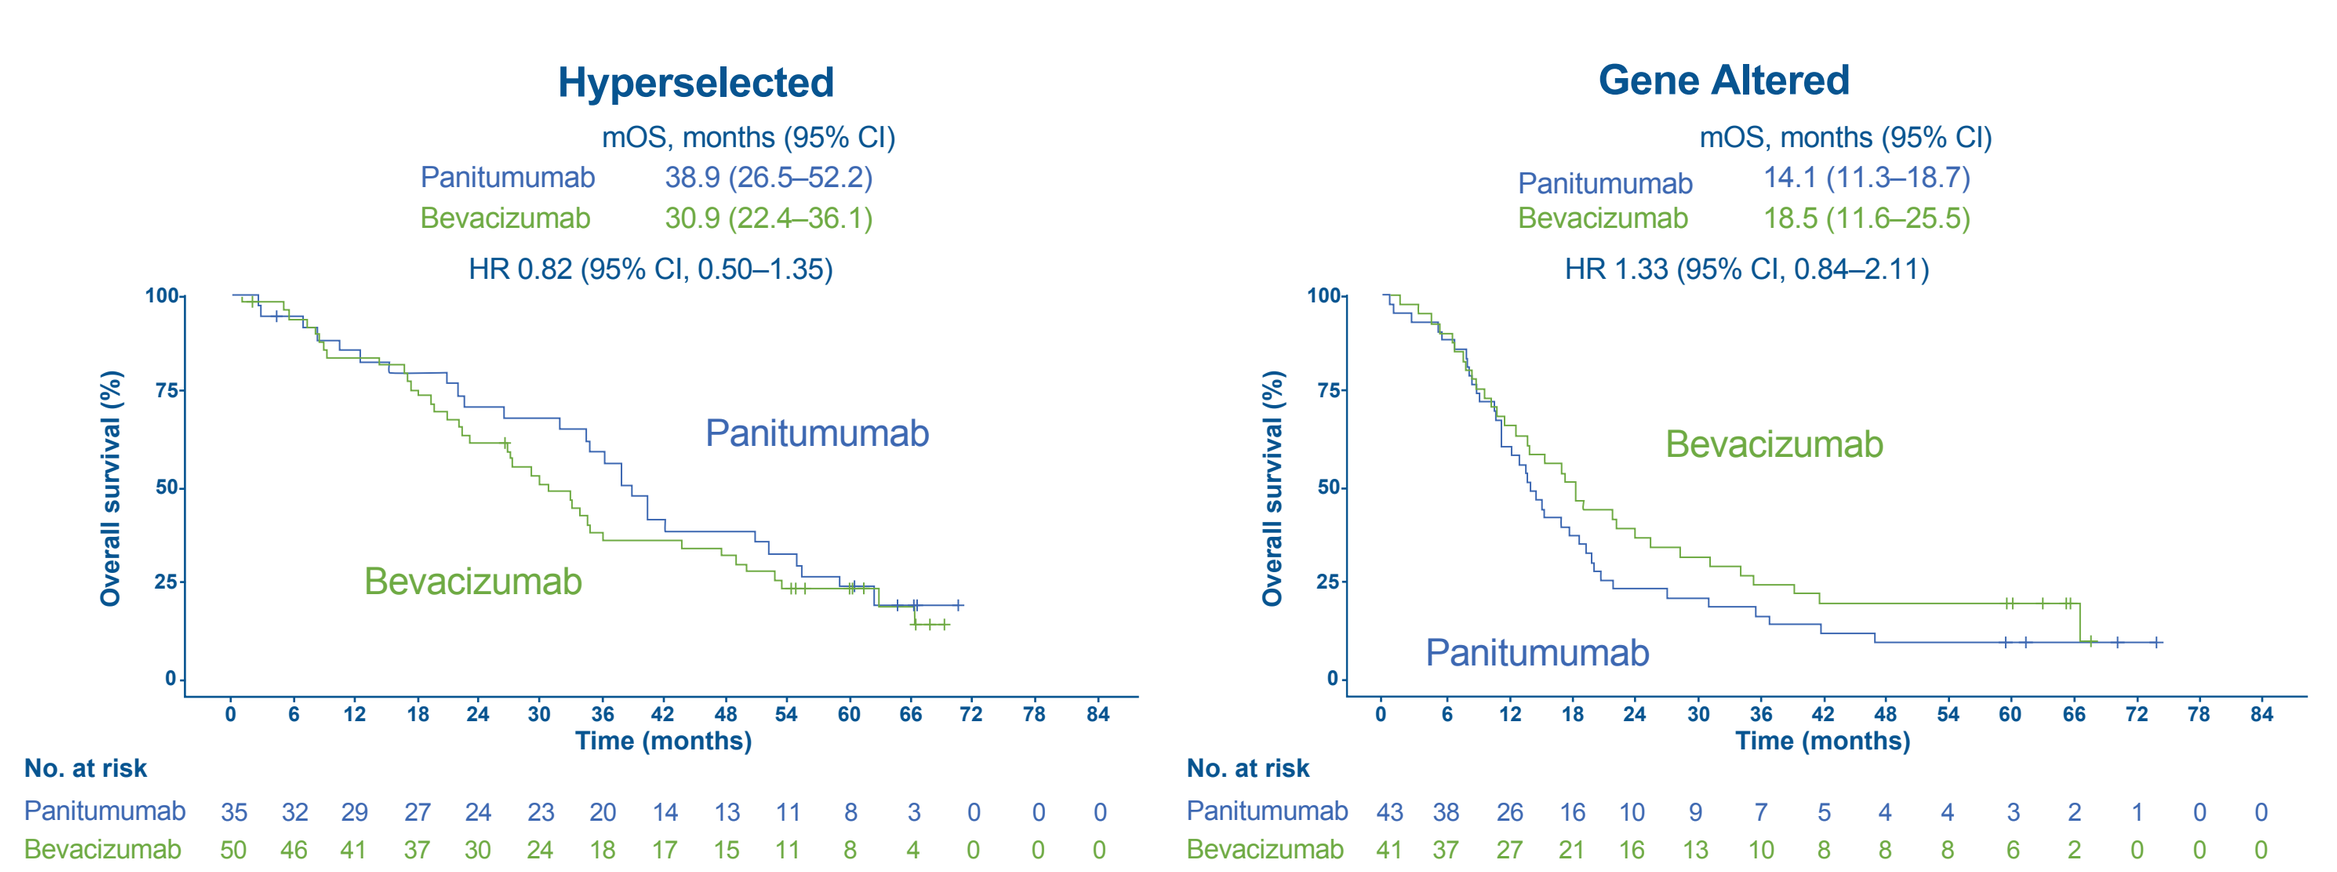
Hyperselected
mOS, months (95% CI)
Panitumumab 38.9 (26.5–52.2)
Bevacizumab 30.9 (22.4–36.1)
HR 0.82 (95% CI, 0.50–1.35)
Gene Altered
mOS, months (95% CI)
Panitumumab 14.1 (11.3–18.7)
Bevacizumab 18.5 (11.6–25.5)
HR 1.33 (95% CI, 0.84–2.11)
Overall survival (%)
Time (months)
Panitumumab
Bevacizumab
No. at risk
Panitumumab 35 32 29 27 24 23 20 14 13 11 8 3 0 0 0
Bevacizumab 50 46 41 37 30 24 18 17 15 11 8 4 0 0 0
No. at risk
Panitumumab 43 38 26 16 10 9 7 5 4 4 3 2 1 0 0
Bevacizumab 41 37 27 21 16 13 10 8 8 8 6 2 0 0 0

Figure 7: Overall survival in ctDNA-analyzed population

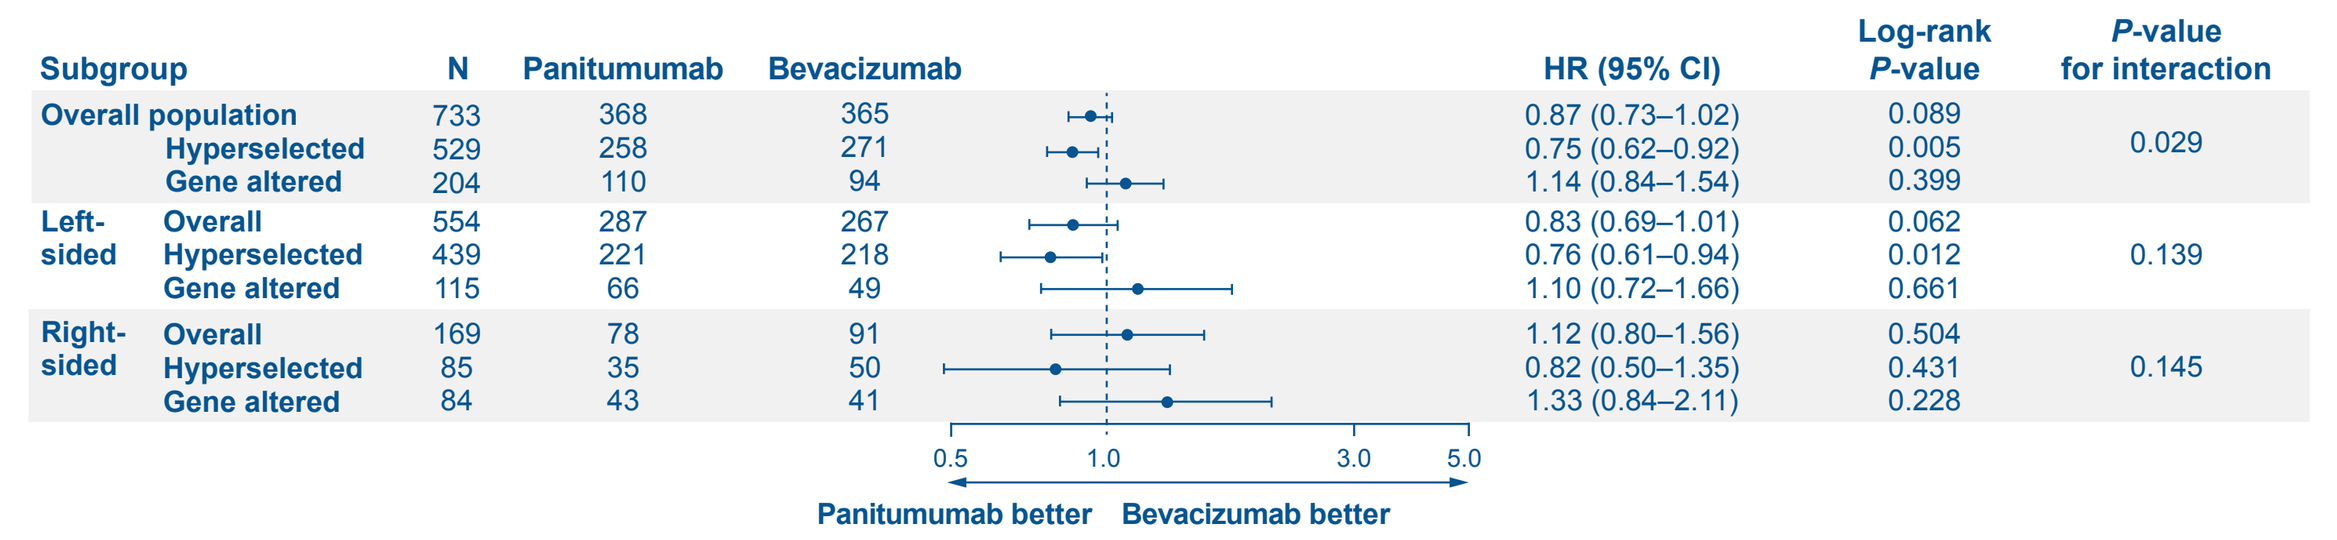
Panitumumab better
Bevacizumab better

Subgroup		N	Panitumumab	Bevacizumab	HR (95% CI)	Log-rank *P*-value	*P*-value for interaction
Overall population		733	368	365	0.87 (0.73–1.02)	0.089	0.029
	Hyperselected	529	258	271	0.75 (0.62–0.92)	0.005	
	Gene altered	204	110	94	1.14 (0.84–1.54)	0.399	
Left-sided	Overall	554	287	267	0.83 (0.69–1.01)	0.062	0.139
	Hyperselected	439	221	218	0.76 (0.61–0.94)	0.012	
	Gene altered	115	66	49	1.10 (0.72–1.66)	0.661	
Right-sided	Overall	169	78	91	1.12 (0.80–1.56)	0.504	0.145
	Hyperselected	85	35	50	0.82 (0.50–1.35)	0.431	
	Gene altered	84	43	41	1.33 (0.84–2.11)	0.228	

Figure 8: Subgroup analysis of overall survival by gene alteration in the overall population analyzed for ctDNA

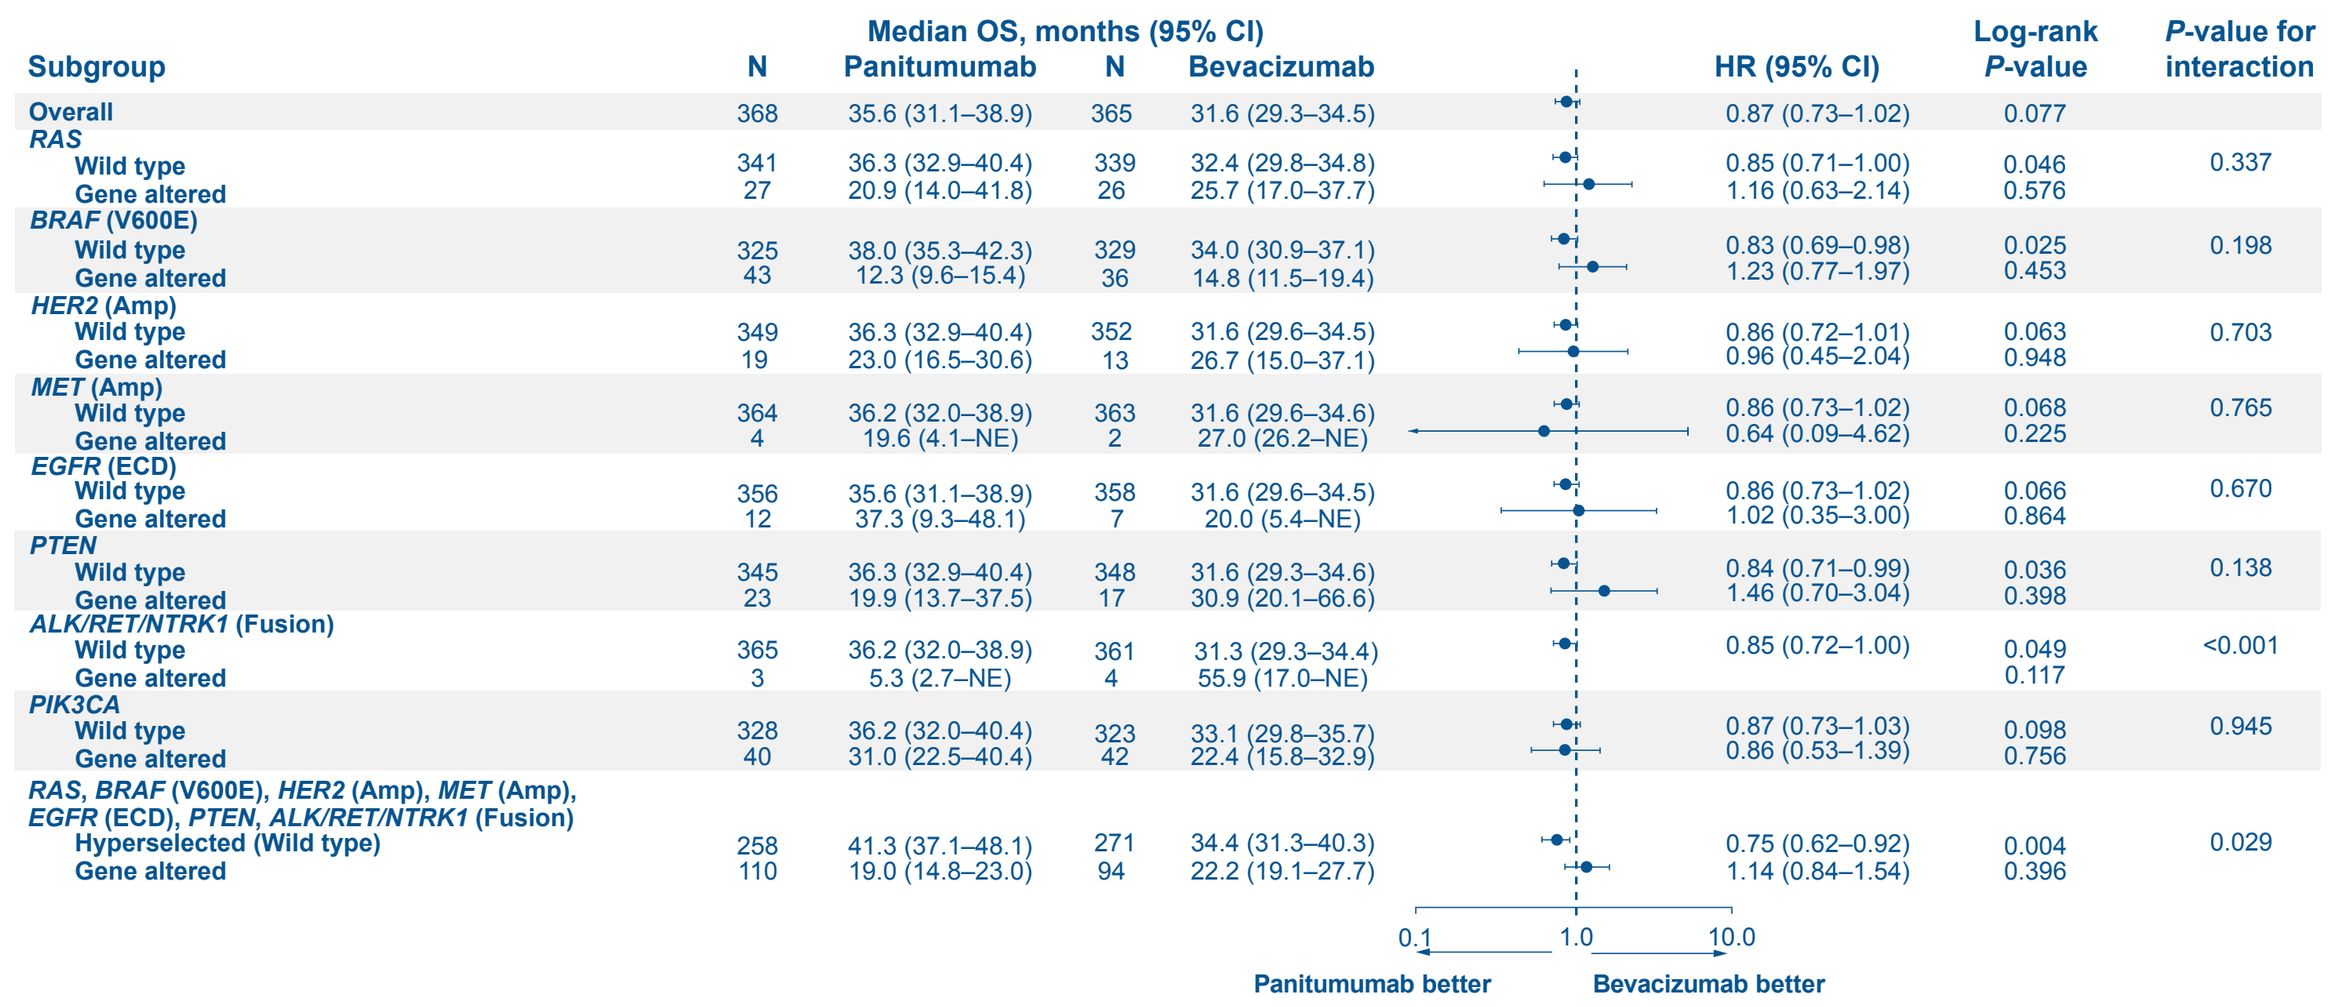
Panitumumab better
Bevacizumab better

Subgroup	N	Median OS, months (95% CI) Panitumumab	N	Bevacizumab	HR (95% CI)	Log-rank *P*-value	*P*-value for interaction
Overall	368	35.6 (31.1–38.9)	365	31.6 (29.3–34.5)	0.87 (0.73–1.02)	0.077	
RAS							
Wild type	341	36.3 (32.9–40.4)	339	32.4 (29.8–34.8)	0.85 (0.71–1.00)	0.046	0.337
Gene altered	27	20.9 (14.0–41.8)	26	25.7 (17.0–37.7)	1.16 (0.63–2.14)	0.576	
BRAF (V600E)							
Wild type	325	38.0 (35.3–42.3)	329	34.0 (30.9–37.1)	0.83 (0.69–0.98)	0.025	0.198
Gene altered	43	12.3 (9.6–15.4)	36	14.8 (11.5–19.4)	1.23 (0.77–1.97)	0.453	
HER2 (Amp)							
Wild type	349	36.3 (32.9–40.4)	352	31.6 (29.6–34.5)	0.86 (0.72–1.01)	0.063	0.703
Gene altered	19	23.0 (16.5–30.6)	13	26.7 (15.0–37.1)	0.96 (0.45–2.04)	0.948	
MET (Amp)							
Wild type	364	36.2 (32.0–38.9)	363	31.6 (29.6–34.6)	0.86 (0.73–1.02)	0.068	0.765
Gene altered	4	19.6 (4.1–NE)	2	27.0 (26.2–NE)	0.64 (0.09–4.62)	0.225	
EGFR (ECD)							
Wild type	356	35.6 (31.1–38.9)	358	31.6 (29.6–34.5)	0.86 (0.73–1.02)	0.066	0.670
Gene altered	12	37.3 (9.3–48.1)	7	20.0 (5.4–NE)	1.02 (0.35–3.00)	0.864	
PTEN							
Wild type	345	36.3 (32.9–40.4)	348	31.6 (29.3–34.6)	0.84 (0.71–0.99)	0.036	0.138
Gene altered	23	19.9 (13.7–37.5)	17	30.9 (20.1–66.6)	1.46 (0.70–3.04)	0.398	
ALK/RET/NTRK1 (Fusion)							
Wild type	365	36.2 (32.0–38.9)	361	31.3 (29.3–34.4)	0.85 (0.72–1.00)	0.049	<0.001
Gene altered	3	5.3 (2.7–NE)	4	55.9 (17.0–NE)		0.117	
PIK3CA							
Wild type	328	36.2 (32.0–40.4)	323	33.1 (29.8–35.7)	0.87 (0.73–1.03)	0.098	0.945
Gene altered	40	31.0 (22.5–40.4)	42	22.4 (15.8–32.9)	0.86 (0.53–1.39)	0.756	
RAS, *BRAF* (V600E), *HER2* (Amp), *MET* (Amp), *EGFR* (ECD), *PTEN*, *ALK/RET/NTRK1* (Fusion)							
Hyperselected (Wild type)	258	41.3 (37.1–48.1)	271	34.4 (31.3–40.3)	0.75 (0.62–0.92)	0.004	0.029
Gene altered	110	19.0 (14.8–23.0)	94	22.2 (19.1–27.7)	1.14 (0.84–1.54)	0.396	

NE, not estimable

Figure 9: Progression-free survival in ctDNA-analyzed population

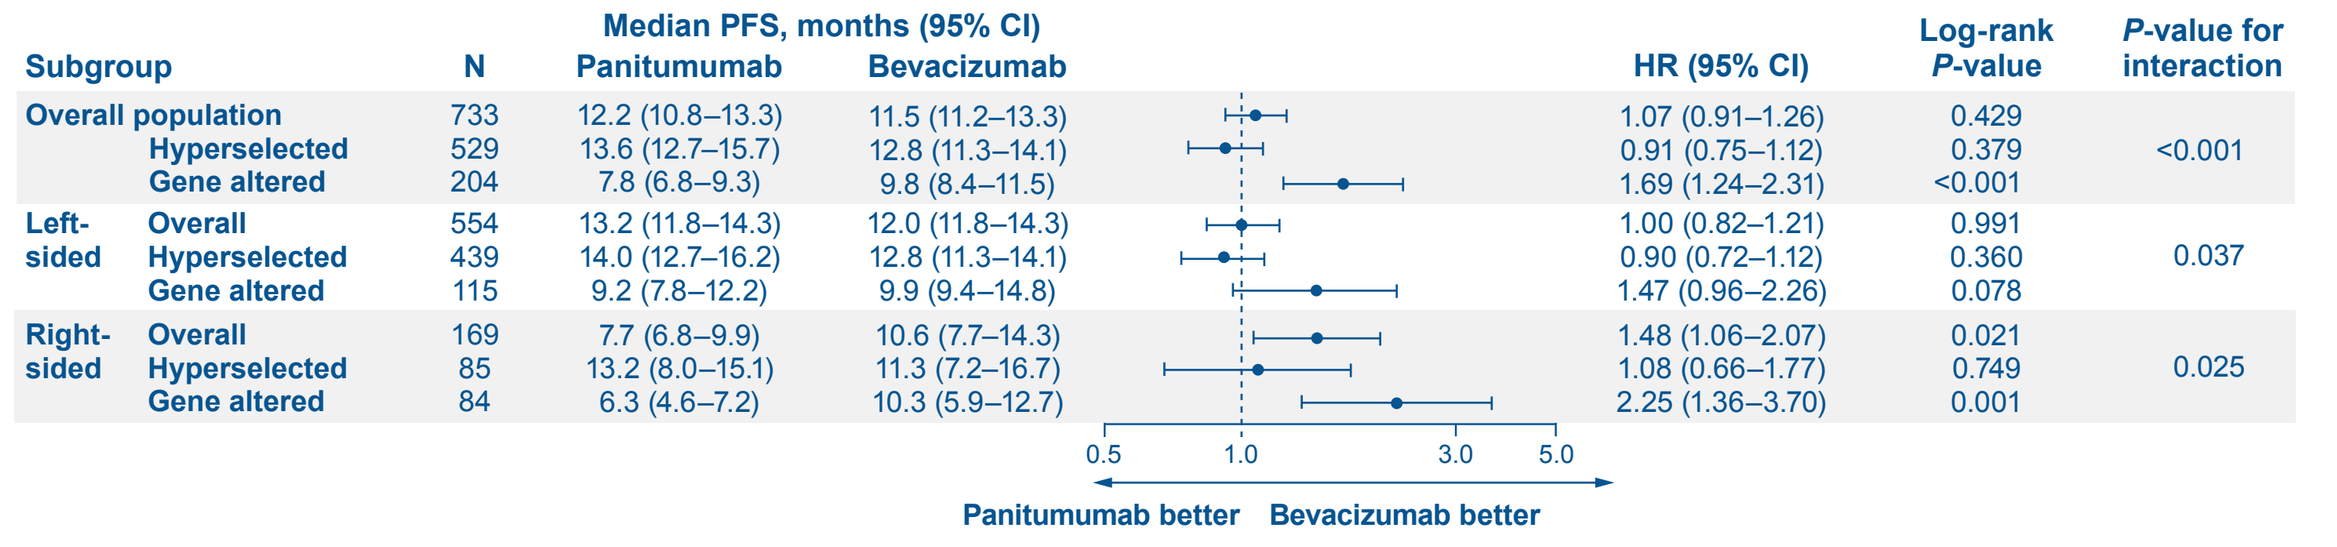
Panitumumab better
Bevacizumab better

Subgroup		N	Median PFS, months (95% CI) Panitumumab	Bevacizumab	HR (95% CI)	Log-rank *P*-value	*P*-value for interaction
Overall population		733	12.2 (10.8–13.3)	11.5 (11.2–13.3)	1.07 (0.91–1.26)	0.429	<0.001
	Hyperselected	529	13.6 (12.7–15.7)	12.8 (11.3–14.1)	0.91 (0.75–1.12)	0.379	
	Gene altered	204	7.8 (6.8–9.3)	9.8 (8.4–11.5)	1.69 (1.24–2.31)	<0.001	
Left-sided	Overall	554	13.2 (11.8–14.3)	12.0 (11.8–14.3)	1.00 (0.82–1.21)	0.991	0.037
	Hyperselected	439	14.0 (12.7–16.2)	12.8 (11.3–14.1)	0.90 (0.72–1.12)	0.360	
	Gene altered	115	9.2 (7.8–12.2)	9.9 (9.4–14.8)	1.47 (0.96–2.26)	0.078	
Right-sided	Overall	169	7.7 (6.8–9.9)	10.6 (7.7–14.3)	1.48 (1.06–2.07)	0.021	0.025
	Hyperselected	85	13.2 (8.0–15.1)	11.3 (7.2–16.7)	1.08 (0.66–1.77)	0.749	
	Gene altered	84	6.3 (4.6–7.2)	10.3 (5.9–12.7)	2.25 (1.36–3.70)	0.001	

Figure 10: Response rate in ctDNA-analyzed population

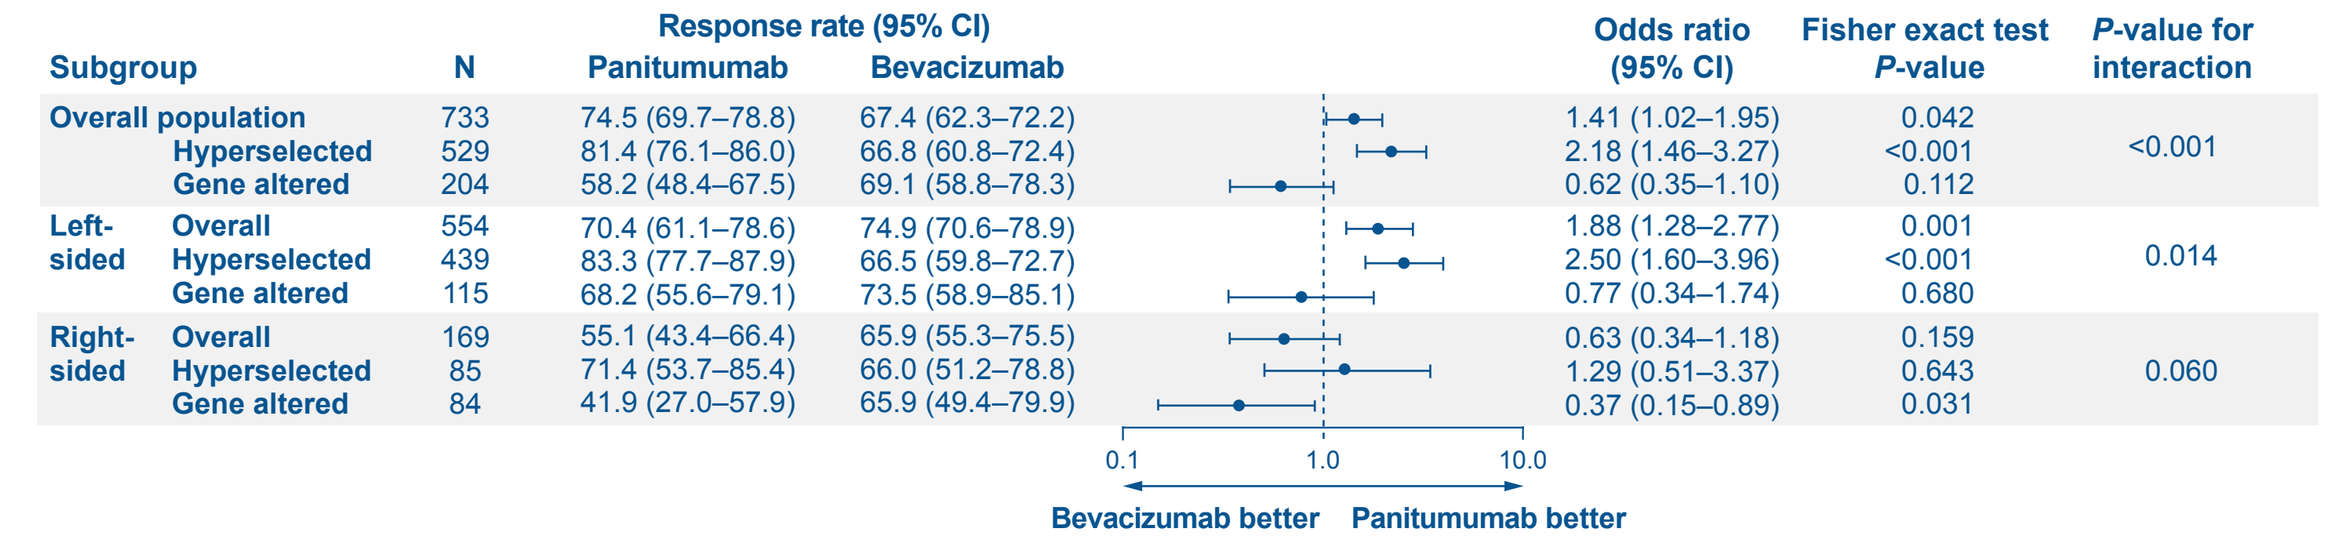
Bevacizumab better
Panitumumab better

Subgroup		N	Response rate (95% CI) Panitumumab	Bevacizumab	Odds ratio (95% CI)	Fisher exact test *P*-value	*P*-value for interaction
Overall population		733	74.5 (69.7–78.8)	67.4 (62.3–72.2)	1.41 (1.02–1.95)	0.042	<0.001
	Hyperselected	529	81.4 (76.1–86.0)	66.8 (60.8–72.4)	2.18 (1.46–3.27)	<0.001	
	Gene altered	204	58.2 (48.4–67.5)	69.1 (58.8–78.3)	0.62 (0.35–1.10)	0.112	
Left-sided	Overall	554	70.4 (61.1–78.6)	74.9 (70.6–78.9)	1.88 (1.28–2.77)	0.001	0.014
	Hyperselected	439	83.3 (77.7–87.9)	66.5 (59.8–72.7)	2.50 (1.60–3.96)	<0.001	
	Gene altered	115	68.2 (55.6–79.1)	73.5 (58.9–85.1)	0.77 (0.34–1.74)	0.680	
Right-sided	Overall	169	55.1 (43.4–66.4)	65.9 (55.3–75.5)	0.63 (0.34–1.18)	0.159	0.060
	Hyperselected	85	71.4 (53.7–85.4)	66.0 (51.2–78.8)	1.29 (0.51–3.37)	0.643	
	Gene altered	84	41.9 (27.0–57.9)	65.9 (49.4–79.9)	0.37 (0.15–0.89)	0.031	

Conclusions

- In hyperselected patients with no gene alterations, OS tended to be longer with panitumumab than with bevacizumab regardless of primary tumor sidedness
 - Overall: mOS, 41.3 vs 34.4 months; HR, 0.75 (95% CI: 0.62–0.92)
 - Left-sided: mOS, 42.1 vs 35.5 months; HR, 0.76 (95% CI: 0.61–0.94)
 - Right-sided: mOS, 38.9 vs 30.9 months; HR, 0.82 (95% CI: 0.50–1.35)
- OS was similar or inferior with panitumumab vs bevacizumab regardless of the primary tumor sidedness in patients with any of these gene alterations
- Negative hyperselection using ctDNA analysis rather than tumor sidedness may identify appropriate patients for first-line panitumumab over bevacizumab
- These results warrant further validation in additional cohorts

References

1. Yoshino T, et al. J Clin Oncol 2022;40(17 suppl):LBA1.
2. Douillard J-Y, et al. N Engl J Med 2013:369:1023–34.
3. Douillard J-Y, et al. J Clin Oncol 2010;28:4697–705.
4. Peeters M, et al. Clin Cancer Res 2013;19:1902–12.
5. Mohan S, et al. PLoS Genet 2014;10:e1004271.

Acknowledgments

We thank the patients, their families, and their caregivers; the PARADIGM investigators and their team members at each study site; and colleagues from Takeda Pharmaceutical Company Limited. Professional medical writing was provided by Kalpana Vijayan, PhD, and Lauren Gallagher, RPh, PhD, of Peloton Advantage, LLC, an OPEN Health company, Parsippany, NJ, USA, and funded by Millennium Pharmaceuticals, Inc., Cambridge, MA, USA, a wholly owned subsidiary of Takeda Pharmaceutical Company Limited. The study was sponsored by Takeda Pharmaceutical Company Limited.

Disclosures

KS: Honoraria (Bristol Myers Squibb, Takeda, and Janssen), consulting/advisory role (Eli Lilly, Bristol Myers Squibb, Takeda, Pfizer, Ono Pharmaceutical, Merck Sharp & Dohme, Taiho, Novartis, AbbVie, GSK, Daiichi Sankyo, Boehringer Ingelheim, Janssen, Amgen, Astellas Pharma, and Guardant Health Japan), research funding, all to institution (Merck Sharp & Dohme, Daiichi Sankyo, Taiho, Chugai Pharma, Ono Pharmaceutical, Astellas Pharma, Medi Science, Eisai, and Amgen); **KM:** Consulting/advisory role (Chugai Pharma, Astra Zeneca, Ono Pharmaceutical, and Amgen), honoraria (Chugai Pharma, Ono Pharmaceutical, Takeda, Eli Lilly, Bayer, Sanofi, Bristol Myers Squibb, and Taiho Pharmaceutical), research grant (Taiho Pharmaceutical, Astellas Pharma, Amgen Astellas Biopharma [rest to institution], Merck Sharp & Dohme, Daiichi Sankyo, Shionogi, Kyowa Kirin, Gilead Sciences, Merck Serono, Pfizer, Sanofi, PAREXEL, Mediscience Planning, Sumitomo Dainippon Pharma, Solasia Pharma, and Ono Pharmaceutical); **JW:** Speakers bureau (Covidien Japan, Johnson & Johnson/Janssen, and Eli Lilly Japan), research funding to institution (Medtronic and TERUMO); **K Yamazaki:** Honoraria (Chugai Pharma, Daiichi Sankyo, Yakult Honsha, Takeda, Bayer, Merck Serono, Taiho Pharmaceutical, Eli Lilly, Sanofi, Ono Pharmaceutical, Merck Sharp & Dohme, and Bristol Myers Squibb), research funding to institution (Taiho Pharmaceutical); **HO:** No relationships to disclose; **MS:** No relationships to disclose; **HY:** No relationships to disclose; **EO:** Speakers bureau (Chugai Pharma, Eli Lilly Japan, Takeda, Ono Pharmaceutical, Bayer Yakuhin, and Bristol Myers Squibb Japan); **TS:** Consulting/advisory role (Takeda), speakers bureau (Chugai Pharma, Eli Lilly Japan, Taiho Oncology, Takeda, Bayer Yakuhin, Ono Yakuhin, and Daiichi Sankyo/UCB Japan); **TN:** Honoraria (Chugai Pharma, Taiho Pharmaceutical, Kaken, Daiichi Sankyo, Eli Lilly Japan, Takeda, Merck, Bayer, and Boehringer Ingelheim), research funding, all to institution (grants from Chugai Pharma, Taiho Pharma, Kaken Pharma, Daiichi Sankyo, and Eli Lilly Japan); **YK:** Speakers bureau (Ono, Taiho, Chugai, Eli Lilly, and Bayer Yakuhin), research funding (Ono, Taiho, Daiichi Sankyo, Chugai, and IQVIA); **TK:** Honoraria (Chugai, Yakult Honsha, Ono Pharmaceutical, Takeda, Eli Lilly Japan, Taiho Pharmaceutical, and Asahi Kasei), research funding (Chugai); **K Yamanaka:** Employment (Takeda); **JS:** Employment (Takeda); **IM:** Employment (Takeda); **MH:** Employment (Takeda); **K Yamamoto:** Honoraria (Chugai, J-Pharma, Johokiko, Triceps, and CMIC Holdings), research funding (Taiho, Boehringer Ingelheim, Takeda, Daiichi Sankyo, and Astellas); **RY:** Honoraria (Chugai Pharma, Takeda, and bitBiome), consulting/advisory role (Takeda); **KA:** No relationships to disclose; **AO:** No relationships to disclose; **HU:** Speakers bureau (Takeda, Chigai, and Taiho); **KT:** Honoraria (Chugai Pharma, Novartis, Takeda, Miyarisan Pharmaceutical, Bristol Myers Squibb, Japan, AstraZeneca, Illumina, Eisai, Boehringer Ingelheim Seiyaku, and Bayer Yakuhin); **TY:** Honoraria (Chugai Pharma, Merck, Bayer Yakuhin, Ono Pharmaceutical, and Merck Sharp & Dohme), research funding, all to institution (Merck Sharp & Dohme, Daiichi Sankyo, Ono Pharmaceutical, Taiho Pharmaceutical, Amgen, Sanofi, Pfizer, Genomedia, Sysmex, Nippon Boehringer Ingelheim, and Chugai Pharma)

Presented at the American Society of Clinical Oncology Gastrointestinal Cancers Symposium, January 19–23, 2023, San Francisco, CA

Email for questions or comments: Kohei Shitara, kshitara@east.ncc.go.jp